A POST HOC ANALYSIS OF SECNIDAZOLE 2 G FOR THE TREATMENT OF BACTERIAL VAGINOSIS: EFFECT OF 2016 FDA GUIDANCE ON CLINICAL RESPONSE RATES IN A SUBPOPULATION OF PATIENTS

Paul Nyirjesy, MD[1]; Joseph L. Amprey, MD, PhD[2]; Maryn Padula, PhD[3]

[1]Drexel University College of Medicine, Philadelphia, PA; [2]Symbiomix Therapeutics, Newark, NJ; [3]Virtuoso Healthcare Group, Manhasset, NY

OBJECTIVES

- The objective of the original study was to evaluate the efficacy and safety of SYM-1219 containing 2 g of secnidazole compared to placebo for the treatment of women and postmenarchal adolescent girls with bacterial vaginosis (BV)
- The purpose of this post hoc analysis was to determine the effects of the updated US Food and Drug Administration (FDA) criteria issued in 2016 on enrollment and clinical response rates

METHODS

Study Design

This was a post hoc analysis from a phase 3, multicenter, prospective, randomized, double-blind placebo-controlled study that assessed the effectiveness and safety of single-dose secnidazole 2 g in women and postmenarchal adolescent girls with BV. All patients or their parental/legal guardians provided written informed consent. Patients were screened for study eligibility at the baseline visit (day 1), and were centrally randomized at a 2:1 ratio to receive either single-dose secnidazole 2 g or matched placebo.

Patients returned to the study center for a visit between study days 7 and 14 and again between study days 21 and 30. Patients could discontinue from the study at any time, for any reason. If patients discontinued due to treatment, an end of study (EOS) visit was conducted and they were offered the option to receive any FDA-approved treatment for BV. The study was conducted in accordance with the International Conference on Harmonization Good Clinical Practice Guideline.

Study Population

Adult females or postmenarchal adolescent girls ≥12 years of age were planned to be enrolled at 21 study centers in the US. All patients had a clinical diagnosis of BV, defined as meeting the following Amsel criteria for BV: discharge; pH ≥4.5, ≥20% clue cells, and positive 10% potassium hydroxide (KOH) whiff test, as well as a Nugent score ≥4 at baseline. For this post hoc analysis, a clinical diagnosis of BV was defined according to the original study, with the added measures that women have a baseline Nugent score ≥7 and a test of cure (TOC) visit occur 7–14 days after administration of the study intervention.[1]

Patients who were pregnant, lactating, menstruating, menopausal, or who were suspected or confirmed clinically of having alternative causes of vaginal symptoms (eg, candidiasis, infections due to *Chlamydia trachomatis*, *Trichomonas vaginalis*, *Neisseria gonorrhoeae*, or genital herpes) were excluded.

Concomitant systemic and topical antimicrobial therapies were not permitted during the course of the study. Alcohol was not permitted for 12 hours prior to study drug administration through 3 days following treatment. Patients were asked to refrain from vaginal intercourse or use of any vaginal products for at least 7 days following treatment.

Outcomes

The primary endpoint assessed in the original modified intent to treat (mITT) study was the proportion of clinical outcome responders (CORs) in the mITT population, defined as those with (1) a normal vaginal discharge, (2) a negative 10% KOH whiff test, and (3) clue cells <20% of the total epithelial cells on microscopic examination of the vaginal wet mount using saline at the TOC/EOS visit (study days 21–30).

An alternate definition was used for the efficacy analysis performed on the mITT population in this post hoc analysis. Using the 2016 FDA criteria, patients in the mITT population with baseline Nugent scores of 7–10 were evaluated for COR rate on days 7–14. Women with an abnormal discharge inconsistent with BV were also considered CORs.

Statistical Analyses

A Cochran-Mantel-Haenszel test adjusted for BV and race strata was used to calculate *P* values. For each treatment group, an exact 95% binomial confidence interval (CI) was calculated for the COR rate.

Sample Size Calculation

Assuming a COR rate of 50% in the single-dose secnidazole 2 g group and 25% in the placebo group, a sample size of 132 patients randomized at a 2:1 ratio provided 80% power to detect a statistically significant difference between single-dose secnidazole 2 g and placebo at a 0.05 level of significance (2-sided). Assuming 75% of patients would have met the mITT criteria, the plan was for 120 patients to be randomized to the single-dose secnidazole 2 g group and 60 patients to the placebo group.

RESULTS

Disposition and Demographics

- A total of 189 patients were enrolled at 21 study centers between May 4, 2015 and August 26, 2015; patients were assigned (2:1) to, and treated with, single-dose secnidazole 2 g (n=125) and placebo (n=64, Figure 1)
- For the original mITT population (the prespecified population for primary analysis), 85.6% (107/125) and 89.1% (57/64) of enrolled patients in the single-dose secnidazole 2 g and placebo groups, respectively, were included:
 - 12 patients were excluded from the mITT population due to a baseline Nugent score <4 (single-dose secnidazole 2 g group, n=10; placebo group, n=2)
 - 13 patients (single-dose secnidazole 2 g group, n=8; placebo group, n=5) were excluded from the mITT population due to a positive sexually transmitted infection test at baseline
- For the updated mITT population based on the 2016 FDA draft guidelines, 71.2% (89/125) and 82.8% (53/64) of enrolled patients in the single-dose secnidazole 2 g and placebo groups, respectively, were included:
 - 22 patients were excluded from the mITT population due to a baseline Nugent score <7 (single-dose secnidazole 2 g group, n=18; placebo group, n=4)

Figure 1. Consort diagram of phase 3 clinical trials of secnidazole vs placebo for treatment of BV

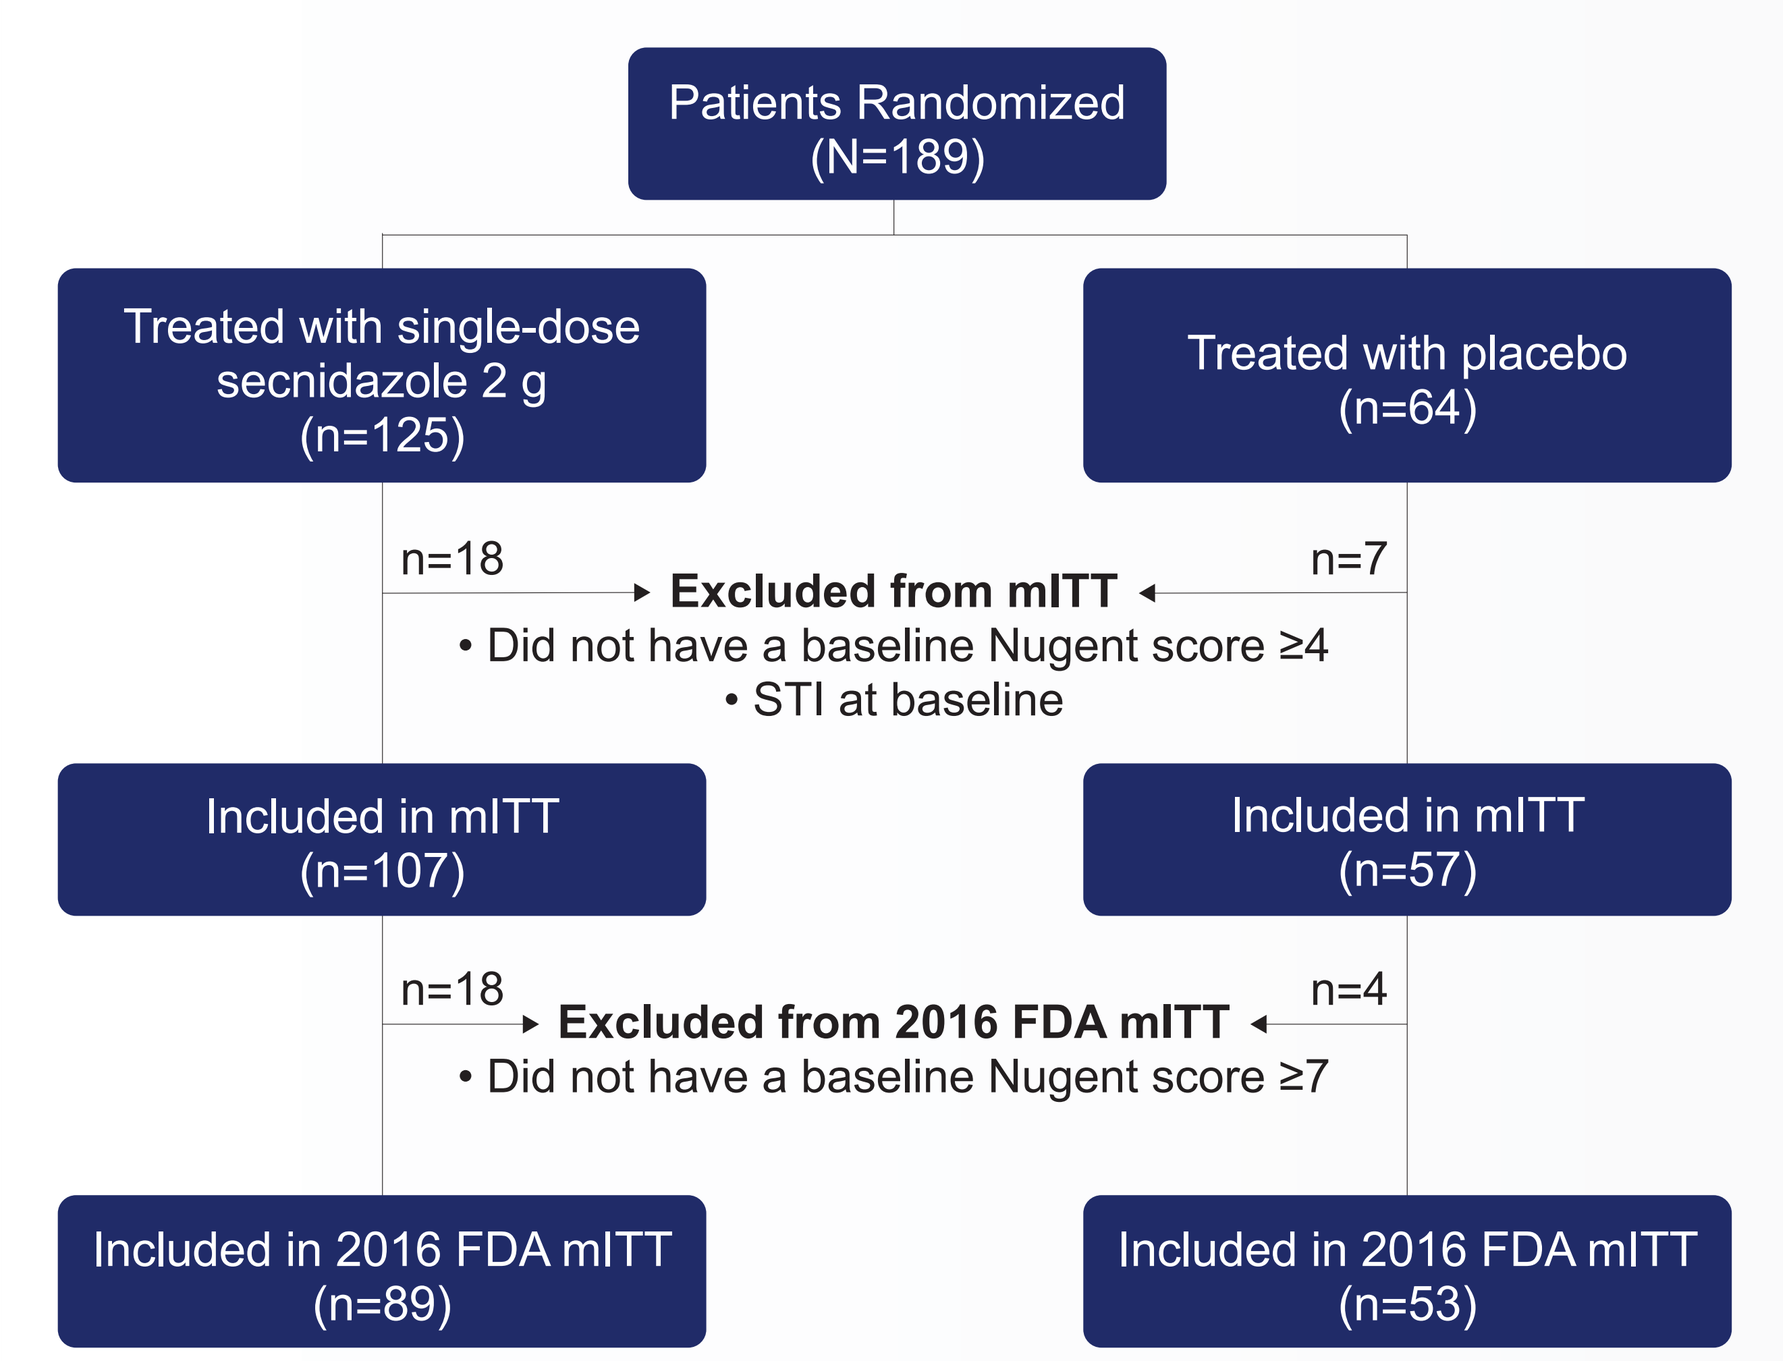
Patients Randomized
(N=189)
Treated with single-dose secnidazole 2 g
(n=125)
Treated with placebo
(n=64)
n=18
Excluded from mITT
n=7
• Did not have a baseline Nugent score ≥4
• STI at baseline
Included in mITT
(n=107)
Included in mITT
(n=57)
n=18
Excluded from 2016 FDA mITT
n=4
• Did not have a baseline Nugent score ≥7
Included in 2016 FDA mITT
(n=89)
Included in 2016 FDA mITT
(n=53)

STI=sexually transmitted infection.

- Among all 164 patients randomized, the mean age was 31 years (range: 18–54 years), 53.7% of patients were black (88/164), and 86.6% had a baseline Nugent score of 7–10 (142/164; Table 1)

Table 1. Demographics and Baseline Characteristics (mITT Population), Overall and by Treatment Group

Parameter	Single-dose Secnidazole 2 g (N=107)	Placebo (N=57)	Overall (N=164)
Age (yr)			
n	107	57	164
Mean (SD)	32 (8.7)	30 (7.6)	31 (8.4)
Median	31	28	30
Min, Max	18, 54	18, 46	18, 54
P value*			0.064
Race, n (%)			
White	46 (43.0)	26 (45.6)	72 (43.9)
Black or African American	59 (55.1)	29 (50.9)	88 (53.7)
Asian	0	1 (1.8)	1 (0.6)
American Indian or Alaska Native	0	0	0
Native Hawaiian or other Pacific Islander	0	0	0
Other	2 (1.9)	1 (1.8)	3 (1.8)
Race strata, n (%)			
Black	59 (55.1)	29 (50.9)	88 (53.7)
All others	48 (44.9)	28 (49.1)	76 (46.3)
P value†			0.625
Number of BV episodes in the past 12 months			
n	107	57	164
Mean (SD)	3 (2.4)	3 (2.6)	3 (2.5)
Median	2	2	2
Min, Max	1, 12	1, 12	1, 12
P value*			0.926
BV strata, n (%)			
≤3 episodes in the past 12 months	83 (77.6)	43 (75.4)	126 (76.8)
≥4 episodes in the past 12 months	24 (22.4)	14 (24.6)	38 (23.2)
P value†			0.846
Nugent score			
n	107	57	164
Mean (SD)	8 (1.8)	9 (1.3)	8 (1.6)
Median	8	8	8
P value*			0.926
Nugent score category, n (%)			
Nugent score 4–6	18 (16.8)	4 (7.0)	22 (13.4)
Nugent score 7–10	89 (83.2)	53 (93.0)	142 (86.6)
P value†			0.095

SD=standard deviation.
**P* value from *t* test for treatment difference.
†*P* value from Fisher exact test for treatment difference.

Efficacy of Secnidazole Based on 2016 Draft Guidance for BV and the Original mITT Study Population

- In the original mITT population, the COR rate for single-dose secnidazole 2 g was superior to placebo (53.3% [57/107; 95% CI (43.4, 63.0)] vs 19.3% [11/57], respectively; *P*<0.001, Figure 2) at the TOC/EOS visit (days 21–30)
- Using the 2016 FDA definition of clinical cure, single-dose secnidazole 2 g was also superior to placebo (64.0% [n=57/89; 95% CI (53.2, 73.9)] vs 26.4% [n=14/53]; respectively, Figure 2) at the interim visit (days 7–14) in the Nugent score 7–10 population

Figure 2. Efficacy based on 2016 FDA draft guidance and the original mITT population

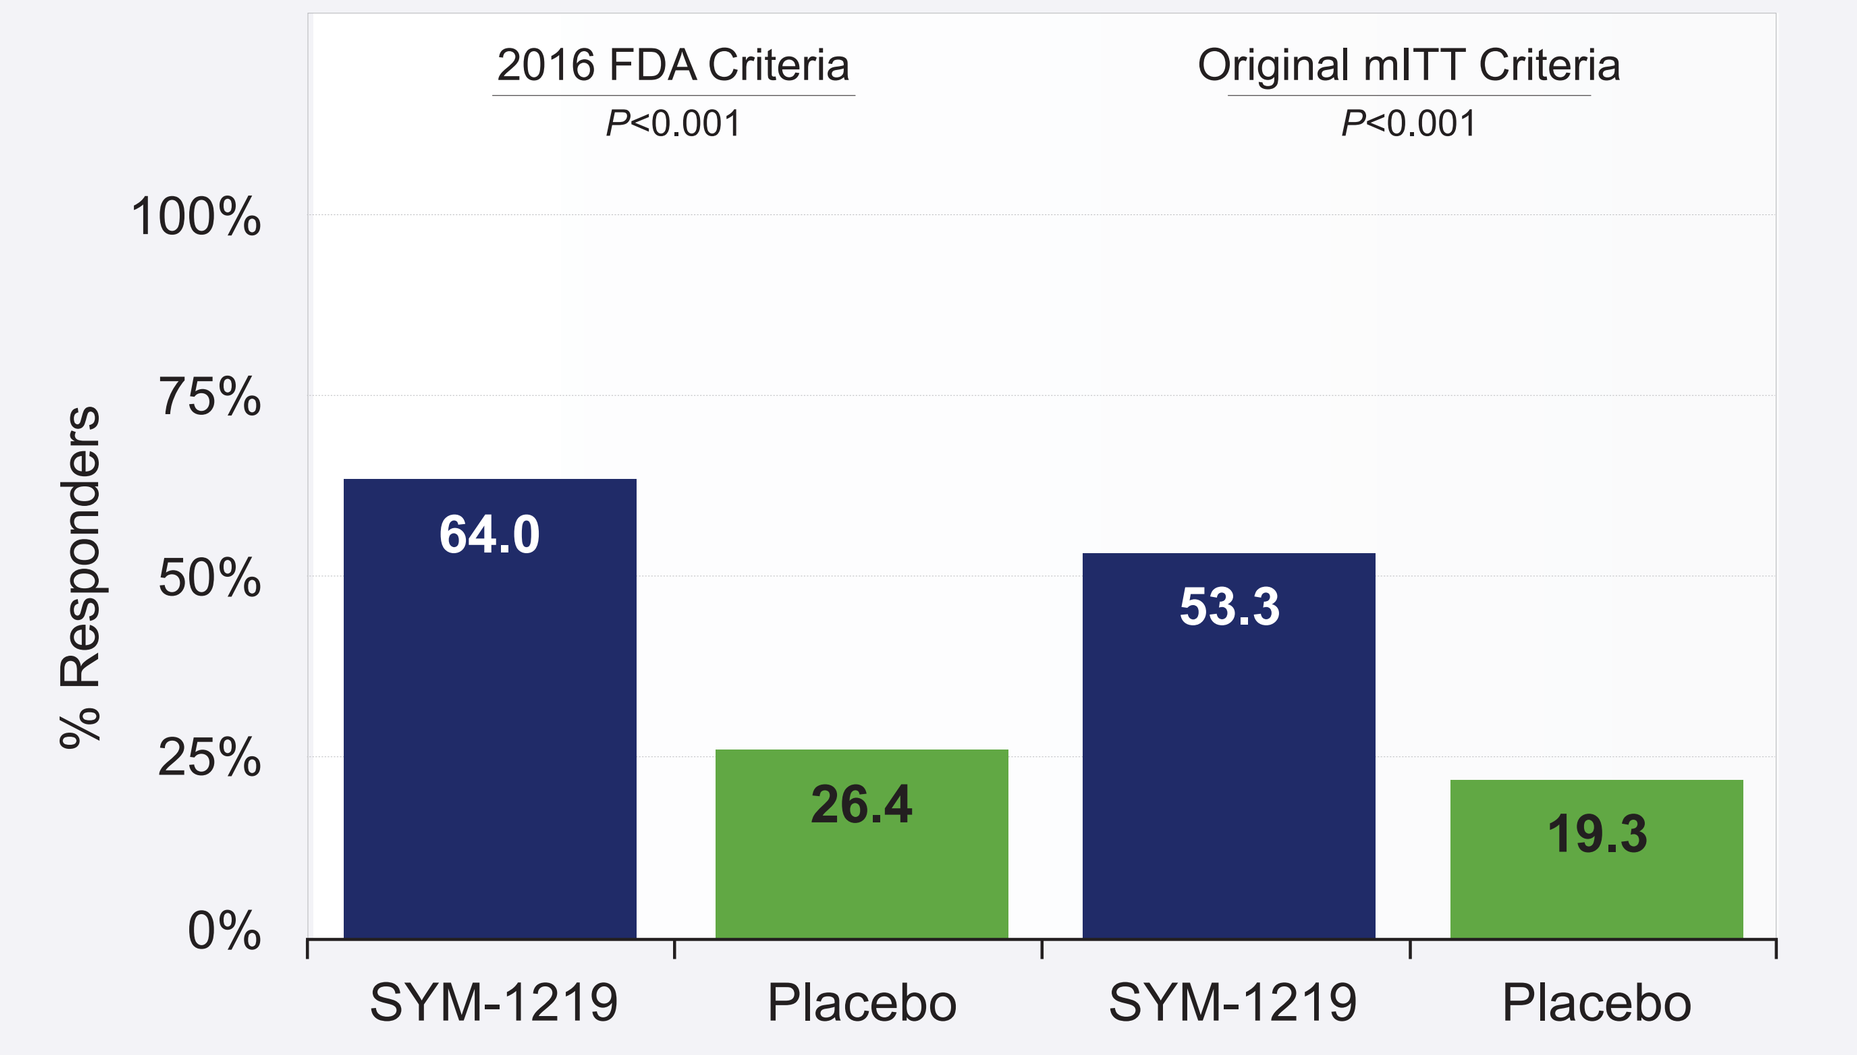
2016 FDA Criteria
P<0.001
Original mITT Criteria
P<0.001
100%
75%
50%
25%
0%
% Responders
64.0
26.4
53.3
19.3
SYM-1219
Placebo
SYM-1219
Placebo

Efficacy of Secnidazole Based on 2016 FDA Draft Guidance at Days 7–14 vs Days 21–30

- Single-dose secnidazole 2 g was superior to placebo (58.4% [n=52/89] vs 24.5% [n=13/53]; *P*<0.001) at the TOC/EOS visit (days 21–30) based on the 2016 FDA clinical cure definition in the Nugent score 7–10 population (Figure 3)
- The response rates at days 7–14 vs days 21–30 were not significantly different (*P*=0.54)

Figure 3. Comparison of efficacy during the interim visit (days 7–14) and the TOC/EOS visit (days 21–30)

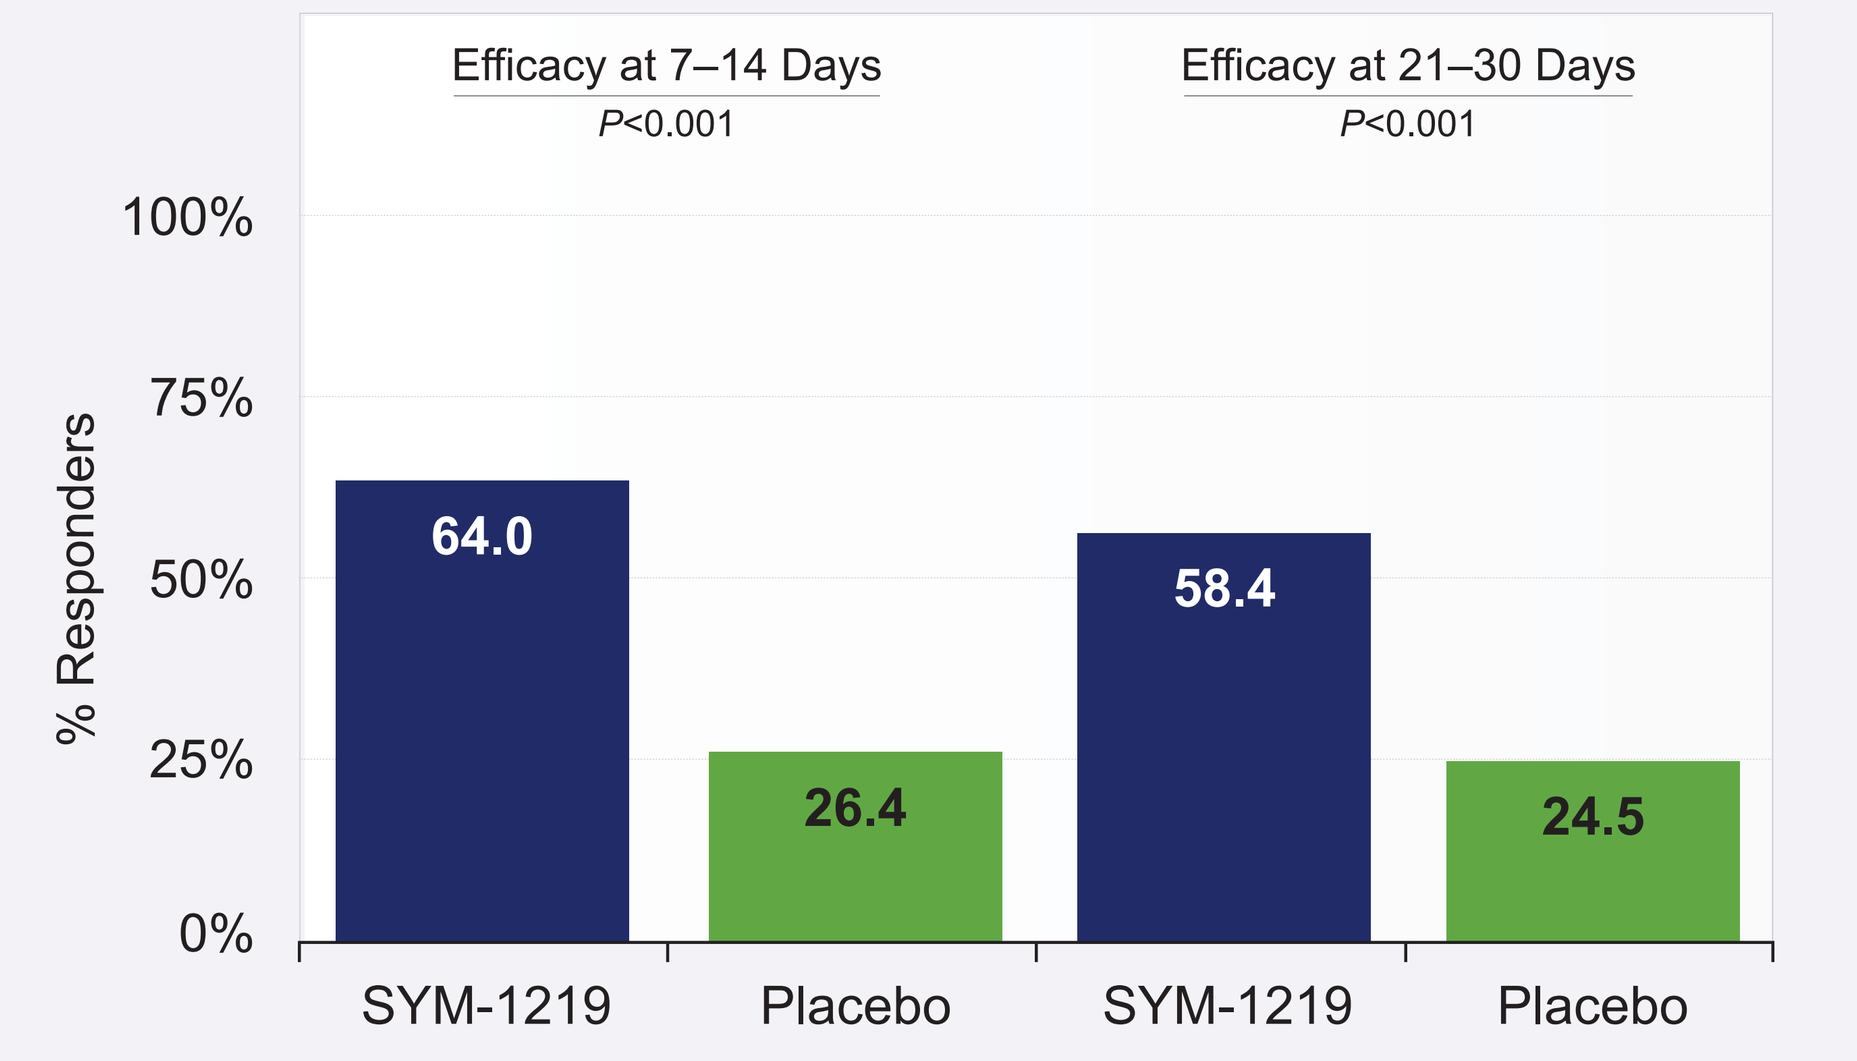
Efficacy at 7–14 Days
P<0.001
Efficacy at 21–30 Days
P<0.001
100%
75%
50%
25%
0%
% Responders
64.0
26.4
58.4
24.5
SYM-1219
Placebo
SYM-1219
Placebo

Efficacy of Secnidazole Based on 2016 FDA Draft Guidance for Severe vs Intermediate BV

- Single-dose secnidazole 2 g had greater efficacy than placebo (77.8% [n=14/18] vs 25.0% [n=1/4]; *P*=0.042) for women with baseline intermediate BV (Nugent score 4–6) assessed at days 7–14 (Figure 4)

Figure 4. Comparison of efficacy in women with Nugent scores 4–6 and 7–10 during the interim visit (days 7–14)

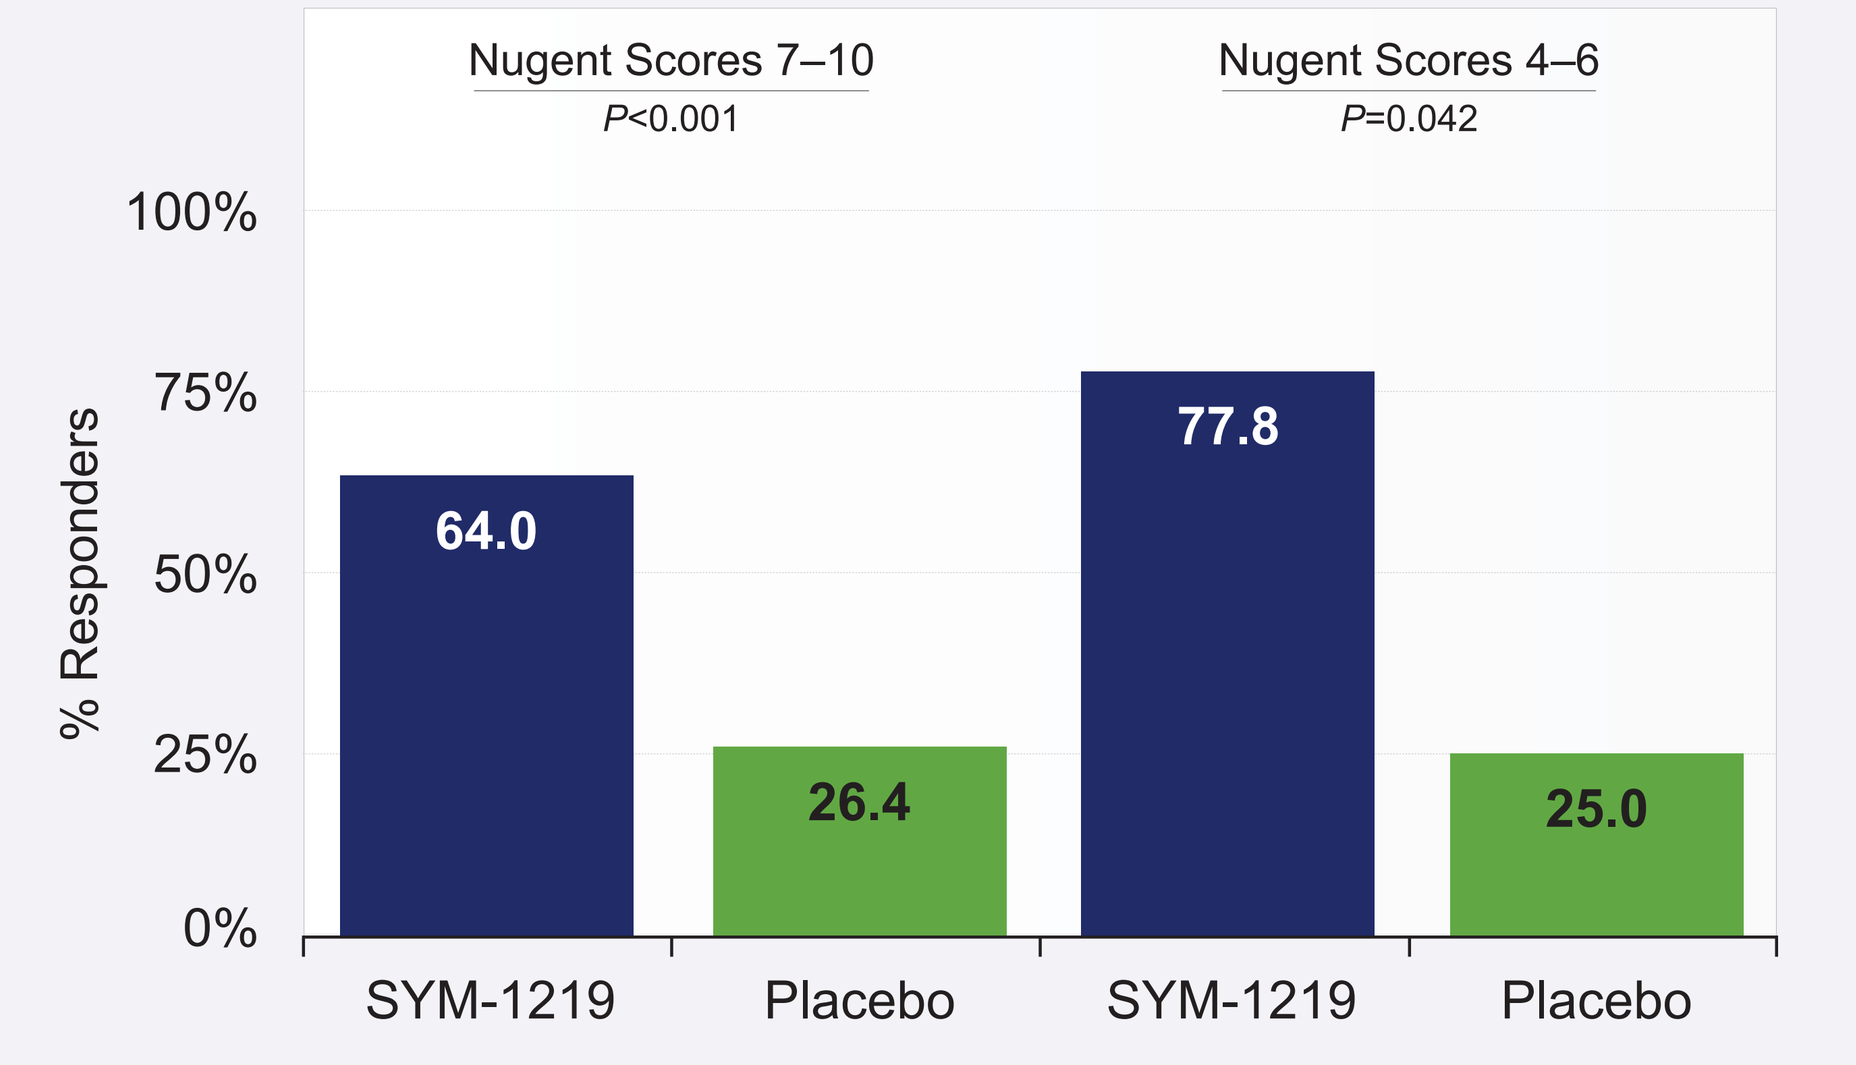
Nugent Scores 7–10
P<0.001
Nugent Scores 4–6
P=0.042
100%
75%
50%
25%
0%
% Responders
64.0
26.4
77.8
25.0
SYM-1219
Placebo
SYM-1219
Placebo

CONCLUSIONS

- Changes between the inclusion criteria for the original mITT analysis and the 2016 draft FDA guidance, particularly Nugent score, excluded a greater number of women
- The COR rate, based on the 2016 draft FDA guidance, determined at days 7–14 vs days 21–30 was not significantly different (*P*=0.54)
- Despite excluding women with BV with intermediate Nugent scores, the COR rate was numerically higher with the 2016 FDA criteria

REFERENCE

1. US Department of Health and Human Services. Food and Drug Administration Center for Drug Evaluation and Research. Bacterial vaginosis: developing drugs for treatment: guidance for industry. https://www.fda.gov/downloads/Drugs/GuidanceComplianceRegulatoryInformation/Guidances/UCM510948.pdf. July 2016. Accessed June 21, 2017.

symbiomix therapeutics