THE EFFECT OF SYM-1219 ON hERG POTASSIUM CHANNELS *IN VITRO* AND ITS IMPLICATIONS ON CARDIAC FUNCTION *IN VIVO*

Borje Darpo, MD, PhD,[1] Nikki Adetoro, MS, MPH,[2] Helen S. Pentikis, PhD[2,3]

[1]iCardiac Technologies, Rochester, NY; [2]Symbiomix Therapeutics, Baltimore, MD; [3]SAJE Consulting, Baltimore, MD

ABSTRACT

Background. SYM-1219, a novel oral granule formulation of secnidazole, an antimicrobial drug in the 5-nitroimidazole class, is under development as a single-dose treatment for bacterial vaginosis. The purpose of these studies was to examine the effects of SYM-1219 on hERG potassium channel current *in vitro* and to evaluate the effect of secnidazole on the QT interval in human subjects.

Methods. *In vitro*, the effect of SYM-1219 (10 or 300 μM) or control was tested on the hERG potassium current using human embryonic kidney cells, and inhibition of the hERG potassium current was measured. In a TQT study, healthy subjects were administered SYM-1219 (2 g or 6 g), placebo, and moxifloxacin in a 4-way crossover design. Serial ECGs were recorded paired with blood samples for PK determination. ECGs were analyzed using the high-precision QT technique. The primary analysis was based on change-from-baseline QTc (ΔQTcF) at postdosing time points, complemented with an exposure response (ER) analysis.

Results. SYM-1219 10 and 300 μM did not inhibit hERG current and IC_{50} could not be determined. Fifty-two subjects were enrolled into the TQT study. The supratherapeutic dose of SYM-1219 (6 g) had a small effect on placebo-corrected ΔQTcF of 7.6 msec (90% CI: 5.5 to 9.7). The slope of the linear ER relationship was shallow with an estimate of 0.058 msec per μg/mL (90% CI: 0.042 to 0.073). Using the ER analysis, a predicted QT effect exceeding 10 msec can be excluded at secnidazole plasma concentrations up to ~125 μg/mL, ~3-fold therapeutic C_{max}.

Conclusion. SYM-1219 does not cause clinically concerning QT prolongation at plasma levels up to 3-fold therapeutic peak concentrations.

OBJECTIVES

- Examine *in vitro* effects of SYM-1219 on the human ether-à-go-go-related gene (hERG) channel current (a surrogate for I_{Kr}, the rapidly activating delayed rectifier cardiac potassium current)[1]
 - » Prolongation of the QT interval has been associated with ventricular arrhythmias[2]
- Demonstrate that SYM-1219 (secnidazole 2 g) has no clinically relevant effect on the QTc interval or other electrocardiogram (ECG) parameters
- Evaluate the relationship between plasma secnidazole concentrations and the effect on QTc interval

symbiomix therapeutics

METHODS

In Vitro *hERG Inhibition Assay*

SYM-1219 was tested for inhibitory effects on the hERG channel current. Human embryonic kidney cells (HEK293) that lack endogenous I_{Kr} were stably transfected with hERG cDNA and transferred to a recording chamber (33°C–35°C). Micropipettes were patch-clamped onto cells, and an amplifier was used for whole-cell recordings.

The vehicle carrier test (SYM-1219) or positive control (terfenadine) solutions (Table 1) were applied to cells held at -80 mV. Onset and steady-state inhibition of hERG potassium current were measured using a pulse pattern with fixed amplitudes (conditioning prepulse +20 mV for 1 second; repolarizing test ramp to -80 mV [-0.5 V/s] repeated at 5 second intervals). Each recording ended with application of a supramaximal concentration of E-4031 500 nM (reference standard), to assess contribution of endogenous current.

Table 1. hERG inhibition assay reagents

Name	Description
Vehicle carrier	HEPES-buffered physiological saline (HB-PS, [pH 7.4]) supplemented with 0.3% (v/v) dimethyl sulfoxide (DMSO) HB-PS: 137 mM NaCl, 4 mM KCl, 1.8 mM $CaCl_2$, 1 mM $MgCl_2$, 10 mM HEPES, 10 mM glucose
Test article	SYM-1219 in HB-PS + 0.3% DMSO • 10 μM • 300 μM
Positive control	Terfenadine 60 nM
Reference standard	E-4031 (Sigma-Aldrich, St. Louis, MO) • 500 nM

hERG Inhibition Data Analysis

Percent inhibition at each concentration of SYM-1219 was compared with vehicle carrier using 1-way ANOVA followed by Dunnett's multiple comparison test. Significant inhibition was defined at the level of $P<0.05$.

TQT Study in Healthy Adult Volunteers

A phase 1, single-center thorough QT (TQT) study assessed the effect of SYM-1219 on ECG parameters in healthy adult male and female subjects. Subjects were randomized to receive a sequence of 4 treatments in this double-blind, placebo-controlled, crossover study (Table 2).

The treatment phase consisted of four 2-day treatment periods with a washout period of 8±2 days between day 1 of each treatment period. A single oral dose of secnidazole 2 g, secnidazole 6 g, placebo, and moxifloxacin 400 mg was given on day 1 in separate periods.

TQT Assessments

Serial ECGs were recorded beginning 1 hour (h) prior to dosing on day 1 through 24 h after dosing. ECGs were extracted in up to 10 replicates/time point, and paired with pharmacokinetics (PK) determination. ECGs were measured with the high precision QT technique.

Blood samples for PK analysis were collected at 30 minutes predosing and serially up to 24 h to determine plasma secnidazole concentrations.

Safety assessments included physical examination, vital signs, clinical laboratory samples, safety ECGs, and recording of adverse events.

Table 2. Treatments administered to healthy volunteers to measure effects of SYM-1219 on TQT intervals

Treatments	
A	1 packet of SYM-1219 oral granules (secnidazole 2 g) with 8 oz of applesauce
B	3 packets, or 6 g, of SYM-1219 oral granules (secnidazole 2 g), with 8 oz of applesauce (supratherapeutic dose)
C	Placebo granules with 8 oz of applesauce
D	Moxifloxacin 400 mg oral tablet

TQT Study Statistical Analysis

The primary analysis for the QT/QTc data was based on change-from-baseline QTc (ΔQTcF) at each of the postdosing time points. Secondary analyses determined effects on other ECG parameters (HR, PR, and QRS). A linear mixed-effects modeling approach determined the relationship between plasma concentration of SYM-1219 and placebo-corrected ΔQTcF (ΔΔQTcF).

RESULTS

hERG Inhibition Assay

- Percent inhibition of the hERG potassium current was similar to vehicle carrier for SYM-1219 at both the 10 μM and 300 μM concentrations
 - » Mean (SEM): 10 μM, 2.9 (0.9%) and 300 μM, 3.0 (1.2%) vs vehicle, 1.6 (0.7%) (Table 3, Figure 1)

Table 3. Percent inhibition on hERG potassium current

Sample and Concentration	Mean	SD	SEM	Number of Cells
Vehicle carrier	1.6%	1.3%	0.7%	3
SYM-1219 10 μM	2.9%	1.7%	0.9%	4
SYM-1219 300 μM	3.0%	2.4%	1.2%	4
Terfenadine 60 nM (positive control)	78.0%	2.7%	NA	2

- Effects of SYM-1219 300 μM on the amplitude of hERG tail currents is represented as a time course in Figure 1
- The IC_{50} for the inhibitory effect of SYM-1219 on hERG potassium current was estimated to be >300 μM

Figure 1. Time course showing effects of SYM-1219 on hERG current

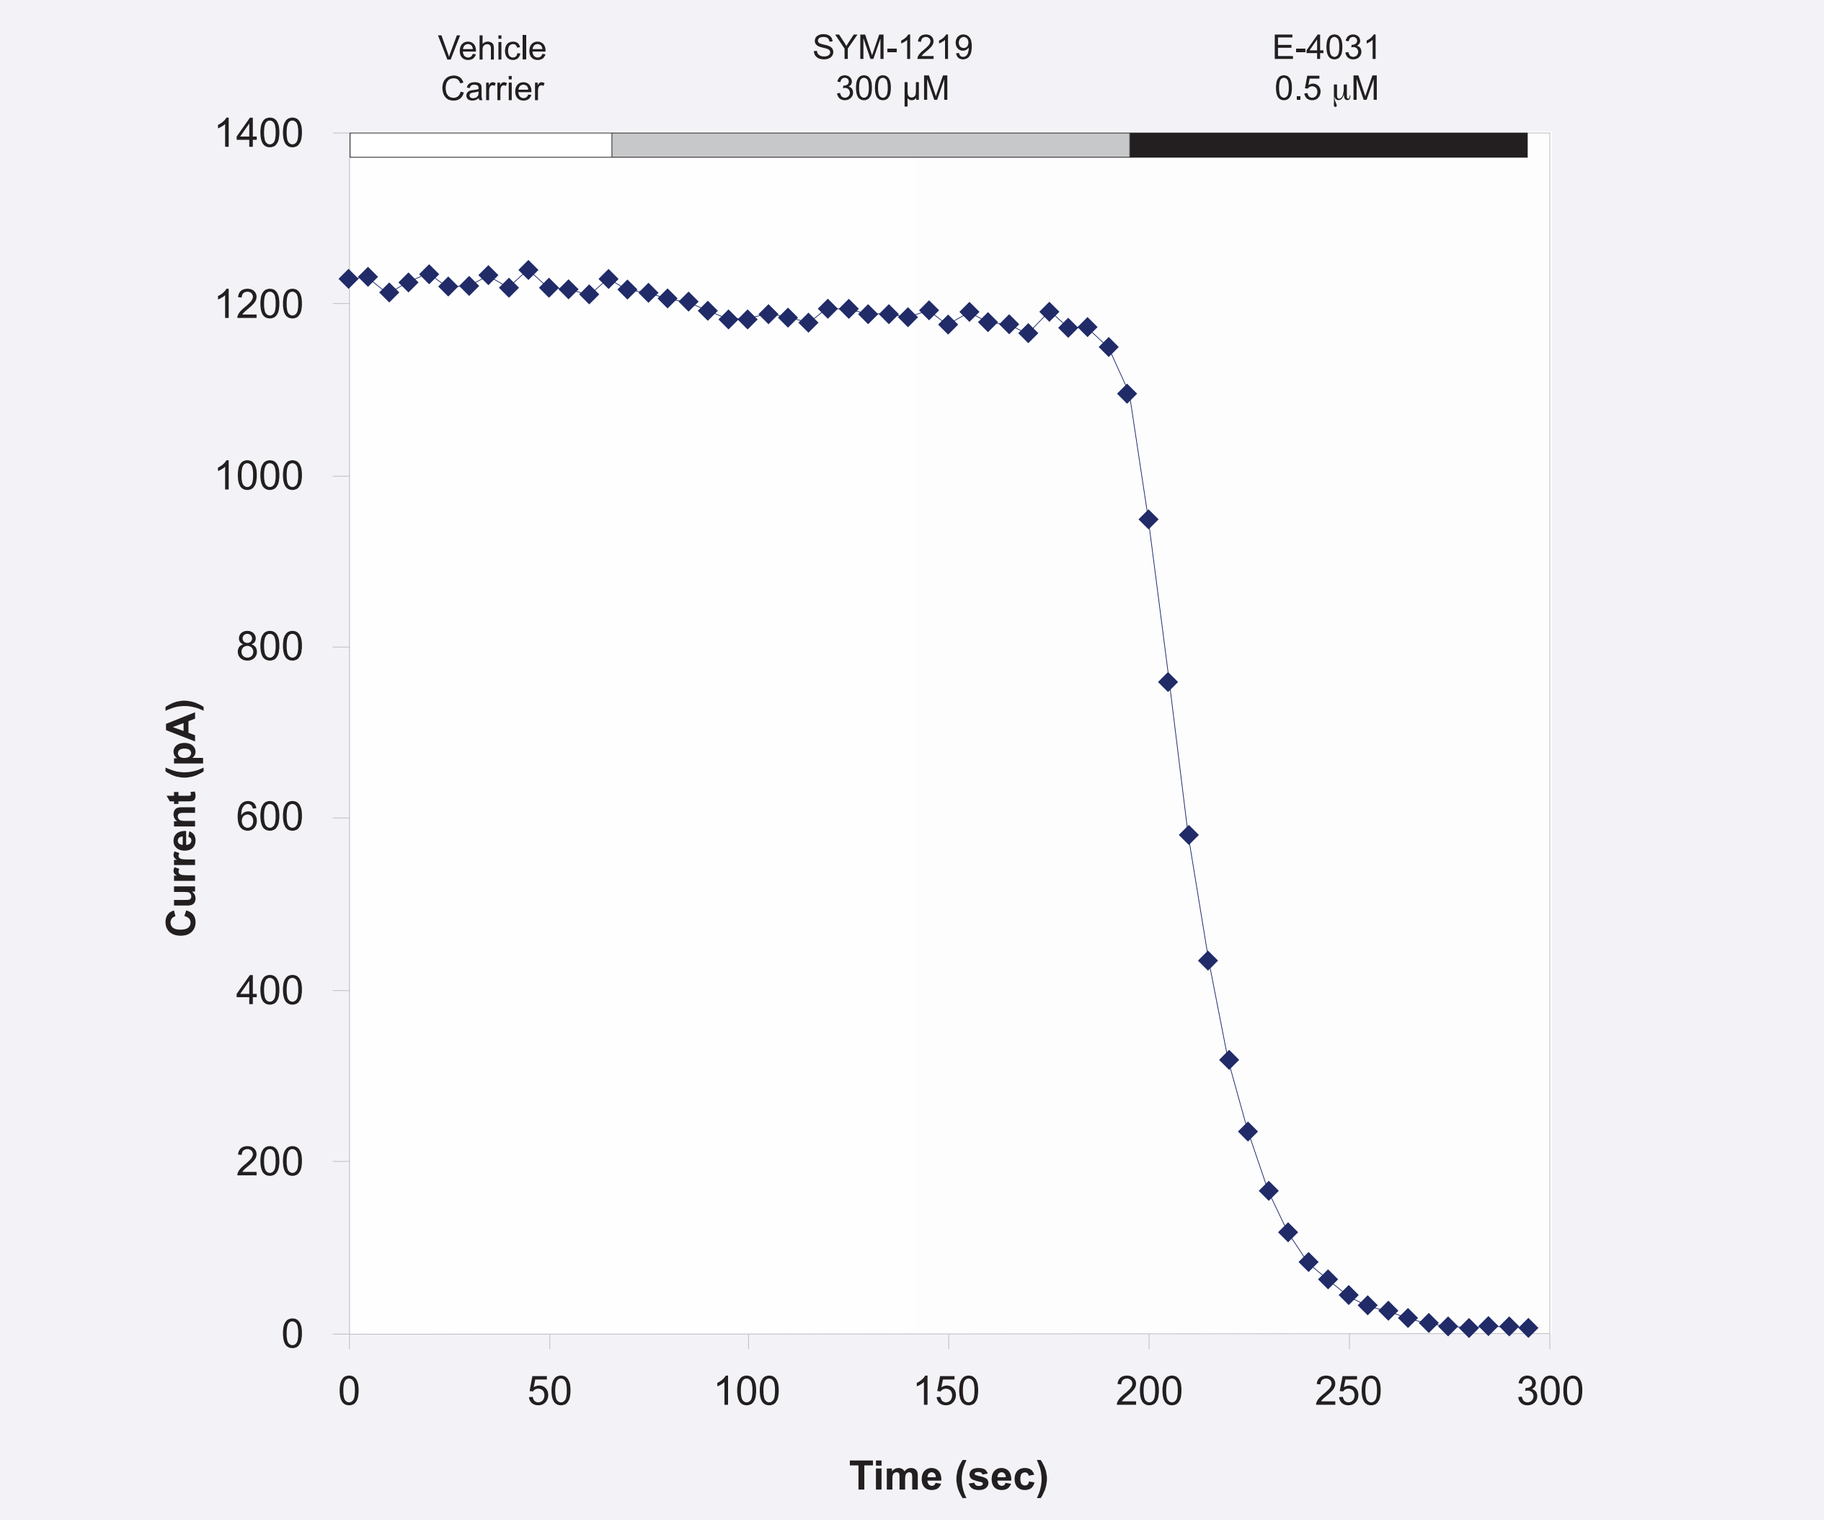

Vehicle Carrier
SYM-1219 300 μM
E-4031 0.5 μM
Current (pA)
Time (sec)

Peak current amplitude during application of vehicle carrier (white bar), test article (gray bar), and reference substance, E-4031 (black bar), is illustrated.

TQT Study in Adult Volunteers

- 52 healthy male (n=30) and female (n=22) subjects were enrolled, and 47 completed the study (Table 4)

Table 4. Baseline patient demographics for the TQT study

Parameter	Overall (N=52)
Age (yr)	
Mean (SD)	45 (11.6)
Gender, n (%)	
Male	30 (57.7)
Female	22 (42.3)
Race, n (%)	
White	45 (86.5)
Black or African American	7 (13.5)

- In the placebo and SYM-1219 2 g dosing periods, mean change-from-baseline QTcF values (ΔQTcF) were negative or close to zero; somewhat larger values were seen from 3 h onward for SYM-1219 6 g
- Placebo-corrected ΔQTcF (ΔΔQTcF) was small with mean values between -0.5 msec and 5 msec after dosing with SYM-1219 2 g at all postdosing time points (Figure 2)
- In the SYM-1219 6 g period, mean ΔΔQTcF was >5 msec between 2 and 8 h postdosing, with the largest values at 4 h (7.6 msec; 90% confidence interval [CI]: 5.5 to 9.7) and 5 h (7.6 msec; 90% CI: 5.5 to 9.7) (Figure 2)
- No QTc outliers were noted with administration of SYM-1219
- Assay sensitivity was demonstrated by the moxifloxacin QT response

Figure 2. Placebo-corrected change-from-baseline QTcF across time point (QT/QTc analysis set)

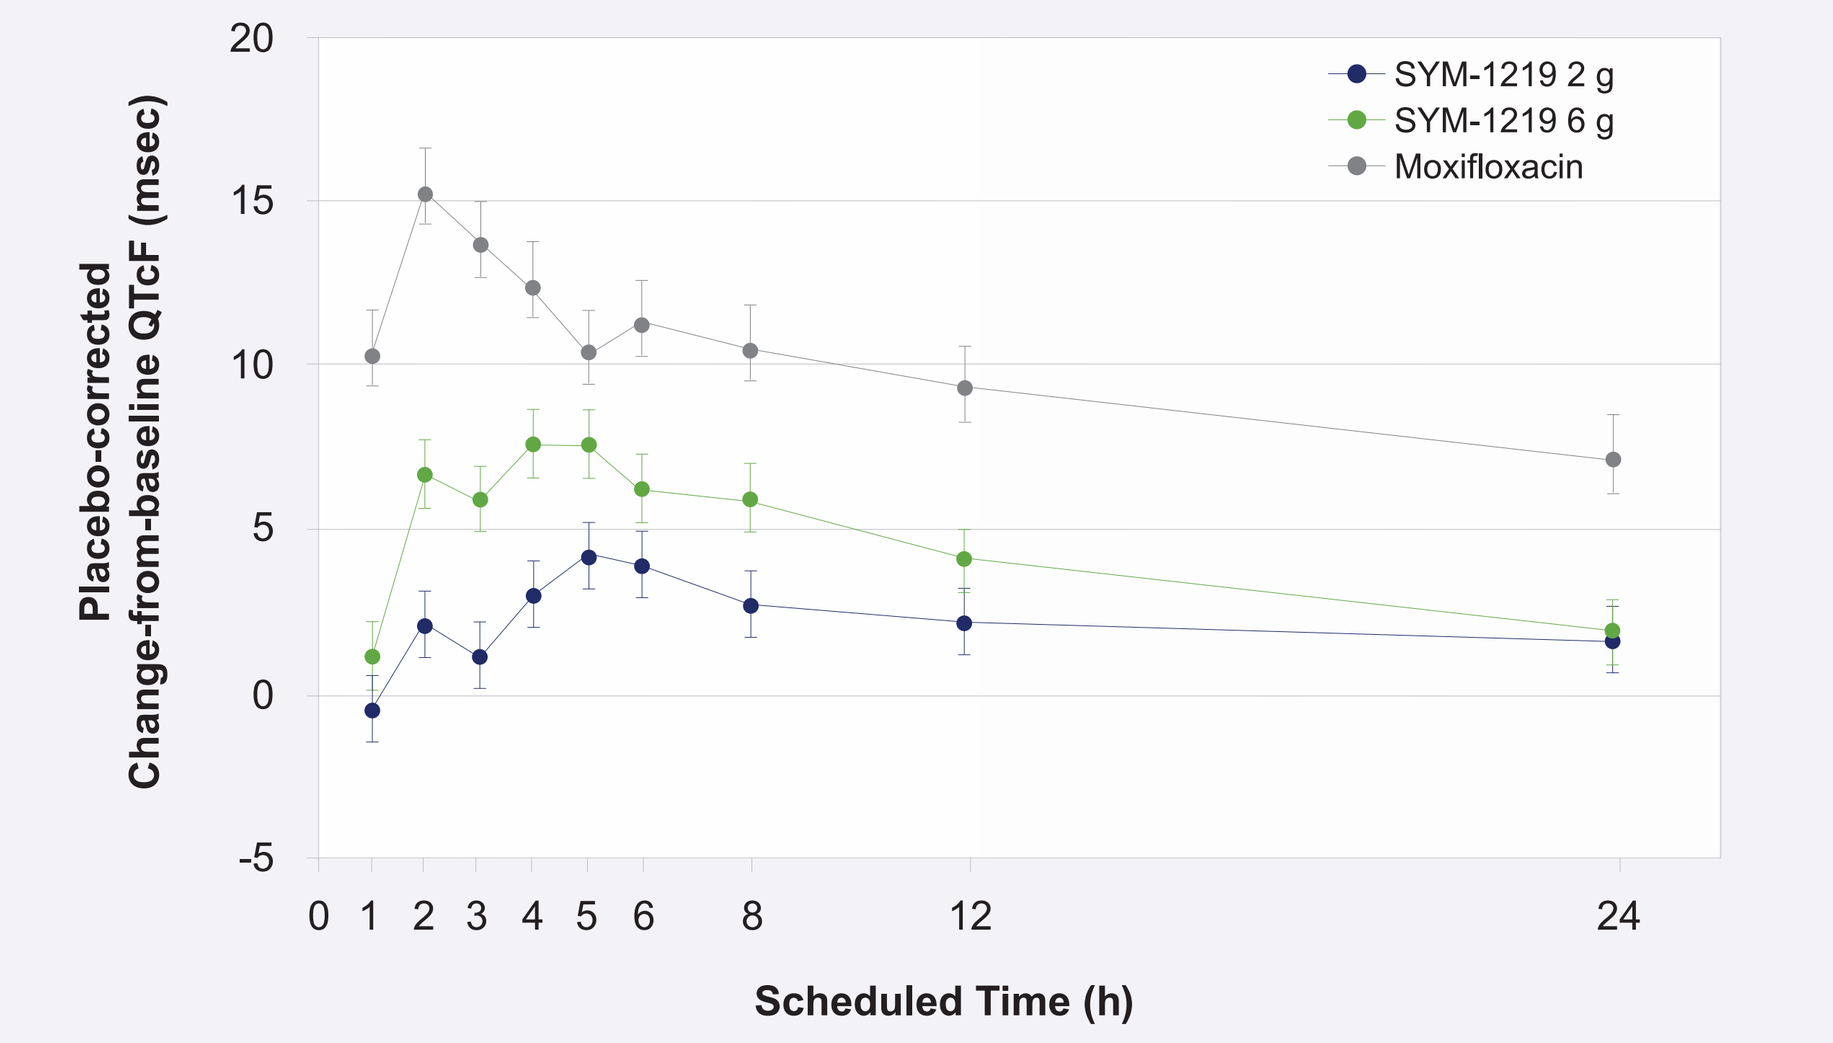

SYM-1219 2 g
SYM-1219 6 g
Moxifloxacin
Placebo-corrected Change-from-baseline QTcF (msec)
Scheduled Time (h)

- No significant safety signals or categorical outliers were noted for other cardiac parameters
- Mean secnidazole plasma concentrations through 24 h are shown in Figure 3
- Dose proportionality was noted for mean maximum plasma concentration (C_{max}): mean [SD]: 2 g, 38.3 (10.4) μg/mL vs 6 g, 113.1 (27.8) μg/mL
- Median T_{max} was slightly longer for the 6 g dose group (5.1 h) vs 2 g dose group (4.1 h)

Figure 3. Mean secnidazole plasma concentrations over time

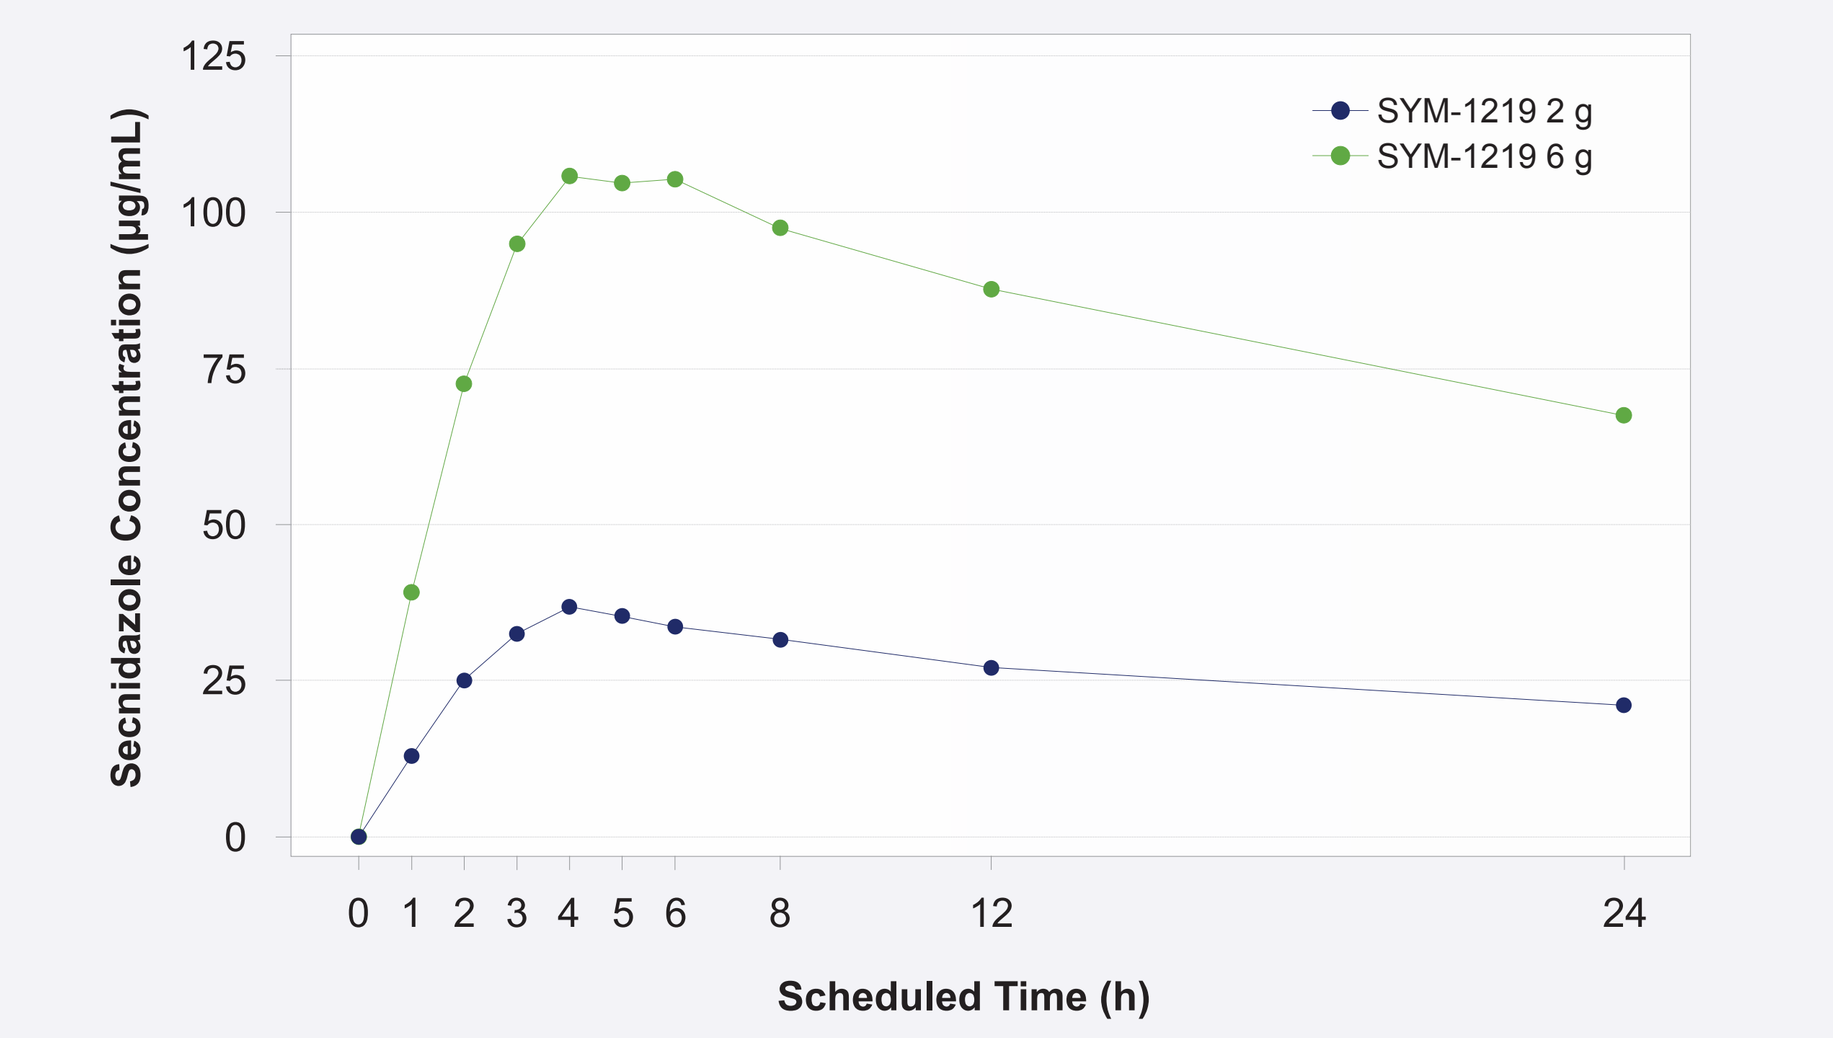

SYM-1219 2 g
SYM-1219 6 g
Secnidazole Concentration (μg/mL)
Scheduled Time (h)

- An exposure response (ER) analysis determined the relationship between SYM-1219 plasma concentration levels and ΔΔQTcF (Figure 3)
 - » The slope of the ER relationship was shallow and statistically significant: 0.058 msec per μg/mL (90% CI: 0.042 to 0.073)
 - » Using the ER relationship, the upper bound of the 90% CI of the predicted QT effect will be below 10 msec at secnidazole plasma concentrations up to ~125 μg/mL, approximately 3-fold greater than therapeutic C_{max}

Figure 4. Model-predicted ΔΔQTcF (mean and 90% CI) and -observed ΔΔQTcF (mean and 90% CI) across deciles of secnidazole plasma concentrations

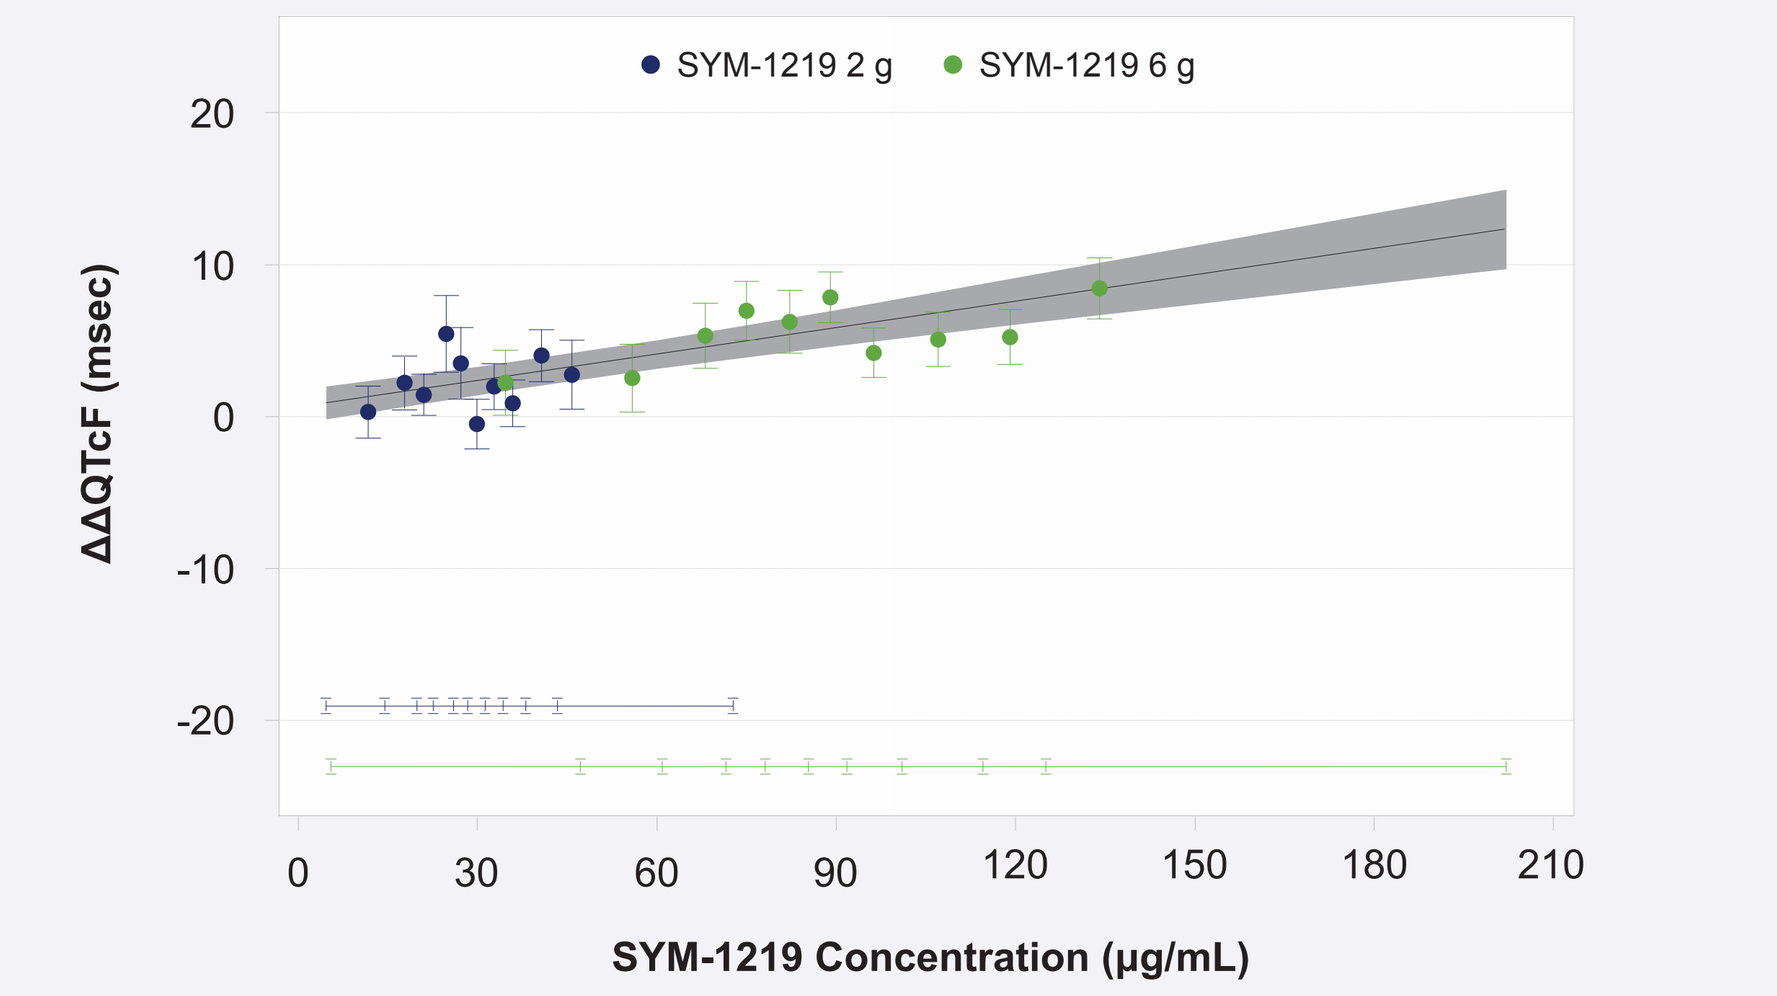

SYM-1219 2 g
SYM-1219 6 g
ΔΔQTcF (msec)
SYM-1219 Concentration (μg/mL)

The solid black line with the gray shaded area shows the model-predicted mean ΔΔQTcF with 90% CI. The blue and green circles with vertical bars are with observed mean ΔΔQTcF with 90% CI at the median secnidazole concentration within each decile. The horizontal blue and green lines with notches show the range of secnidazole concentrations, divided into deciles.

- Treatment-emergent adverse events (TEAEs) were mild to moderate. Incidence of TEAEs was higher in SYM-1219 groups (24.0% and 20.0% for 6 g and 2 g treatments, respectively) compared with controls (8.3%, placebo; 4.3%, moxifloxacin)
- Most frequently reported events: headache, dizziness, and nausea
- One subject discontinued due to a non–treatment-related TEAE (atrial fibrillation) after administration of 2 g SYM-1219

CONCLUSIONS

- The IC_{50} for SYM-129 on hERG potassium current was estimated to be >300 μM, suggesting minimal inhibitory effects
- ER analysis demonstrated that a clinically relevant QT effect can be excluded at secnidazole plasma concentrations up to 125 μg/mL, 3-fold above therapeutic concentrations
- Both doses evaluated in the TQT study were safe and well tolerated
- Taken together, *in vitro* and clinical assessment of the effects of SYM-1219 on cardiac repolarization suggest no clinically meaningful concerns

REFERENCES

1. Redfern WS, Carlsson L, Davis AS, et al. Relationships between preclinical cardiac electrophysiology, clinical QT interval prolongation and torsade de pointes for a broad range of drugs: evidence for a provisional safety margin in drug development. *Cardiovasc Res.* 2003;58(1):32-45.
2. Brown AM, Rampe D. Drug-induced long QT syndrome: is HERG the root of all evil? *Pharmaceutical News.* 2000;7(4):15-20.